Death of Elders Due to Resident-to-Resident Incidents in Dementia in Long-Term Care Homes

Eilon Caspi PhD, School of Nursing, University of Minnesota, Minneapolis, MN

University of Minnesota
Driven to Discover®

Overview

- Resident-to-resident incidents (RRI) in long-term care (LTC) homes are a prevalent, concerning but underrecognized phenomenon (Lachs et al. 2016).
- A growing number of studies examined various aspects of RRI in LTC homes, including prevalence, characteristics, and causes (McDonald et al. 2015).
- One groundbreaking study examined physical injuries caused by RRI in nursing homes (Shinoda-Tagawa et al. 2004).
- Only one study examined fatal RRI in LTC homes in Australia (Murphy et al. 2017).
- **No studies have been conducted on fatal RRI in North America.**

Objectives

- Examine the circumstances surrounding the death of elders as a result of RRI in dementia in LTC homes.
- Identify practically useful patterns to inform prevention.
- The study is *Not* meant to identify the incidence of fatal RRI.

Qualitative Research Methods

Source of data (All publically available information):
- Newspaper articles published online (over 150)
- Death Review Reports (GTLCRC to CCO, 1990-2016)

Comprehensive Internet search: Spring 2012 – Fall 2017

Data detection and extraction: Structured Guide

Data Analysis: Time period: Summer – Fall 2017
- Miles & Huberman (1994) approach
- Qualitative review and abstraction of narratives
- Complemented with tabulation by aggregation / counts
- Simple descriptive statistics

Findings

n = Number of deaths for which data were available

- Identified **105 deaths** of elders (> 60 y/o) as a result of RRI in dementia (at least one of the residents involved in the incident had dementia)
- Time period: Deaths occurred between 1988 - 2017
- Type of LTC home (n=50): Majority in nursing homes; 26% in assisted living
- Countries: Canada (n=51); USA (n=42); Australia & New Zealand (n=4 & n=2); UK (n=5); Singapore (n=1)

Characteristics of Residents
- Age targets (n=103): 84.5 years old (average)
- Age exhibitors (n=76): 75.2 years old (average)
- Gender targets (n=100): Men 52%; Women 48%
- Gender exhibitors (n=99): Men 74%
- Newly admitted residents (< 3 months): 23 deaths

The Circumstances Surrounding the Deaths:

- Location (n=84): Inside bedrooms (59%)
- Time of day (n=63): Evening (44%) and Night (14%)
- Weekend (n=94): 38%
- Roommates (n=77): 43%
- Not witnessed by staff (n=84): 62%
- Nature of physical contact (n=99): "Push-Fall" incidents (44%); Head and/or face beating (22%)
- Object used against target (n=88): 31%
- Nature of physical injury (n=79): Head/face or brain injuries (50%); Hip fractures (33%)
- Cause of death (n=69):
 Blunt head trauma (29%)
 Complications from fractures (20%)
 Pneumonia (11%); Strangulation/Suffocation (10%)
- Time until death (n=95): 16 days (average); 24% died on same day

Practical Implications

The patterns, gaps in supervision, and vulnerability areas identified could inform efforts to prevent deaths in similar circumstances.

This could be accomplished through:

Staff training programs (e.g. recognition, prevention, de-escalation).

Increase staffing levels/supervision during vulnerability time periods.

Strengthen residents' meaningful engagement ("activities") program.

Policies and procedures (e.g. admission; roommates' assignment).

Physical environment (shift to private bedrooms; floor plan/layout).

Develop and use assistive technology (e.g., to alert staff in real time).

Limitations

Incomplete data; Limited ability to verify accuracy of data (such as diagnosis of dementia); small sample limiting generalizability

Future Directions

- Develop a centralized surveillance / medico-legal dataset.
- Conduct the first national study on injurious and fatal RRI (such as using coroner records and police records).
- Bridge gap in MDS 3.0 Section E Behaviors (Caspi, 2013).
- Develop a survey deficiency citation (F-Tag) for RRI in CMS-certified nursing homes (for 20 reasons why, see Caspi, 2017).
- Conduct research in assisted living (Caspi, 2015) & VA LTC homes.
- Evaluate staff training program to demonstrate reduction in RRI.

Acknowledgements

The study was supported by Theresa Piccolo, wife of Frank Piccolo, Toronto, Canada